Water Contamination Risk Assessment for IWRM in Vietnam

Dipl.-Geogr. Sylvia Jaschinski, Dipl.-Ing. Andreas Borgmann, Dipl.-Ing. Nils Führer, Dipl.-Ing. Christian Jolk, Dipl.-Geogr. Sandra Greassidis, Prof. Dr. Harro Stolpe, Dipl.-Geogr. Björn Zindler

Environmental Engineering + Ecology, Faculty of Civil and Environmental Engineering
Ruhr-University of Bochum, Universitaetsstr. 150, 44810 Bochum, Germany
Fon +49 (0)234 32 - 27995, Fax +49 (0)234 32 - 14701
http://www.rub.de/ecology, http://www.iwrm-vietnam.vn, Email: harro.stolpe@rub.de

Summary

The German-Vietnamese joint research project „Integrated Water Resources Management Vietnam" (IWRM-Vietnam) funded by the German Federal Ministry of Education and Research (BMBF) is developing Planning and Decision Support Tools (PDST) for Integrated Water Resources Management in Vietnam.

The Institute of Environmental Engineering and Ecology (eE+E) at the University of Bochum is closely cooperating with the Department of Water Resources Management (DWRM) of the Vietnamese Ministry of National Resources and Environment (MoNRE) in order to develop a method in compliance with Vietnamese conditions (data availability, administration and legislation).

The research focuses on the development and application of GIS based Planning and Decision Support Tools on a river basin scale as well as local technological concepts for sustainable water management in three regions of Vietnam: Upper Dong Nai basin (Province Lam Dong), Red River part basin (Province Nam Dinh), Mekong part basin (City Can Tho) (cf. fig 1).

The approach facilitates a consideration of water resources quantity and quality leading to the identification of Water Management Units (WMUs) with priority need for action. The methodical concept consists of three main tools: **Water Balance Tool**, **Contamination Risk Tool** and **Ranking Tool.** This poster is showing an example of the **Contamination Risk Tool** in chapter "Contamination Risk Tool". Chapter "Results and Ranking" is giving an outlook on the **Ranking Tool** and its use.

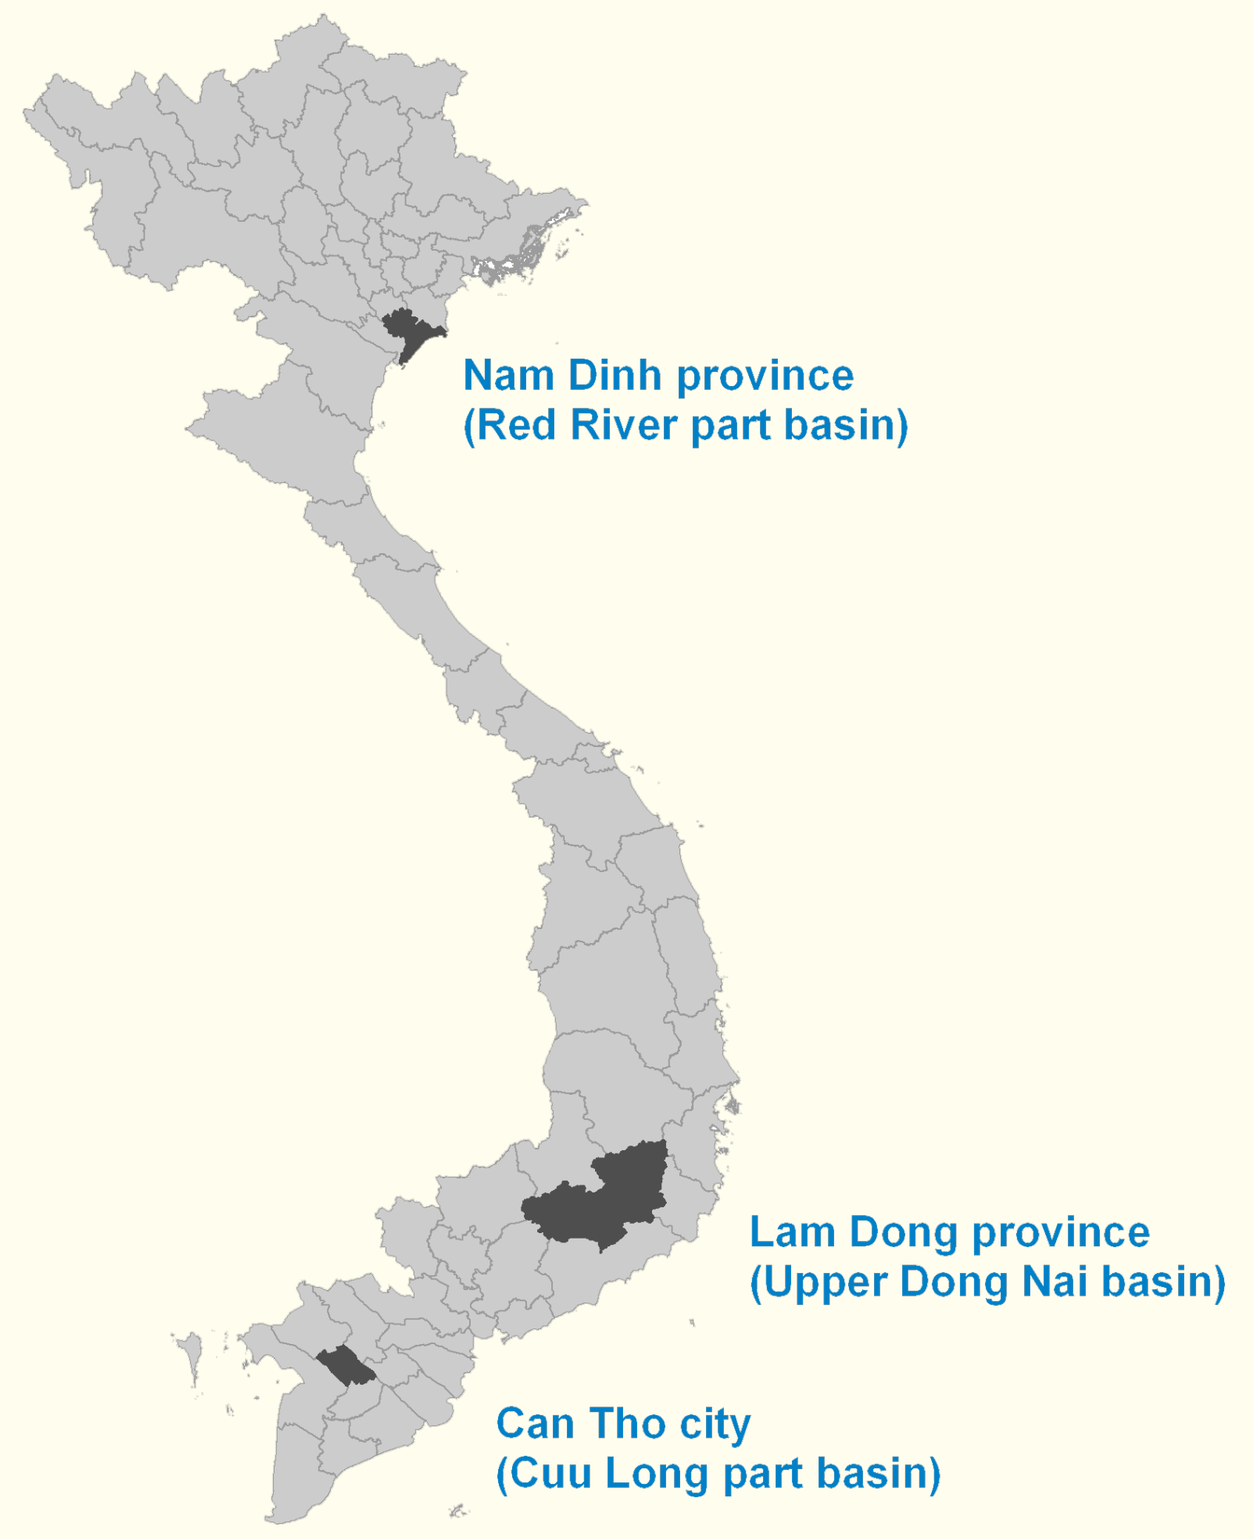
Nam Dinh province
(Red River part basin)
Lam Dong province
(Upper Dong Nai basin)
Can Tho city
(Cuu Long part basin)

Figure 1. Overview of the selected research regions of IWRM-Vietnam

Introduction

Vietnam is a country with rich water resources. A dense river network provides an abundant supply of water. Despite this comfortable situation Vietnam faces numerous challenges:

- The uneven rainfall across Vietnam and the dry seasons result in water supply problems in some areas.
- Vietnam partly is a downstream country. As a consequence, the quantity and quality of the surface water partly depends on the water use in the upstream countries.
- The infrastructure for water supply, wastewater management, flood protection etc. is still deficient and large investments will be required during the coming decades.
- The dynamics of population and economic growth, industrialization, expansion and intensification of agricultural land use and deforestation with all their negative side effects lead to a rapidly increasing water demand and to severe water pollutions. These effects are intensified by an insufficient water supply and sanitation infrastructure.
- Even though Vietnam has recently undertaken many steps to re-organize its institutional and legislative framework ([1-4]), the water sector still is in a state of structural transition.

Project IWRM-Vietnam is part of the beginning implementation process of IWRM principles in Vietnam (as defined by the Global Water Partnership [5]). Project IWRM-Vietnam is embedded in the general framework of five planning levels on different scales. The detail of the planning levels increases from the top to the bottom of the planning pyramid (cf. fig. 2):

1. International level (international guidelines: e.g. GWP)
2. National planning level (national policy, legislation)
3. River basin planning level
4. Water Management Units (WMUs) planning level
5. Local sites planning level (local measures)

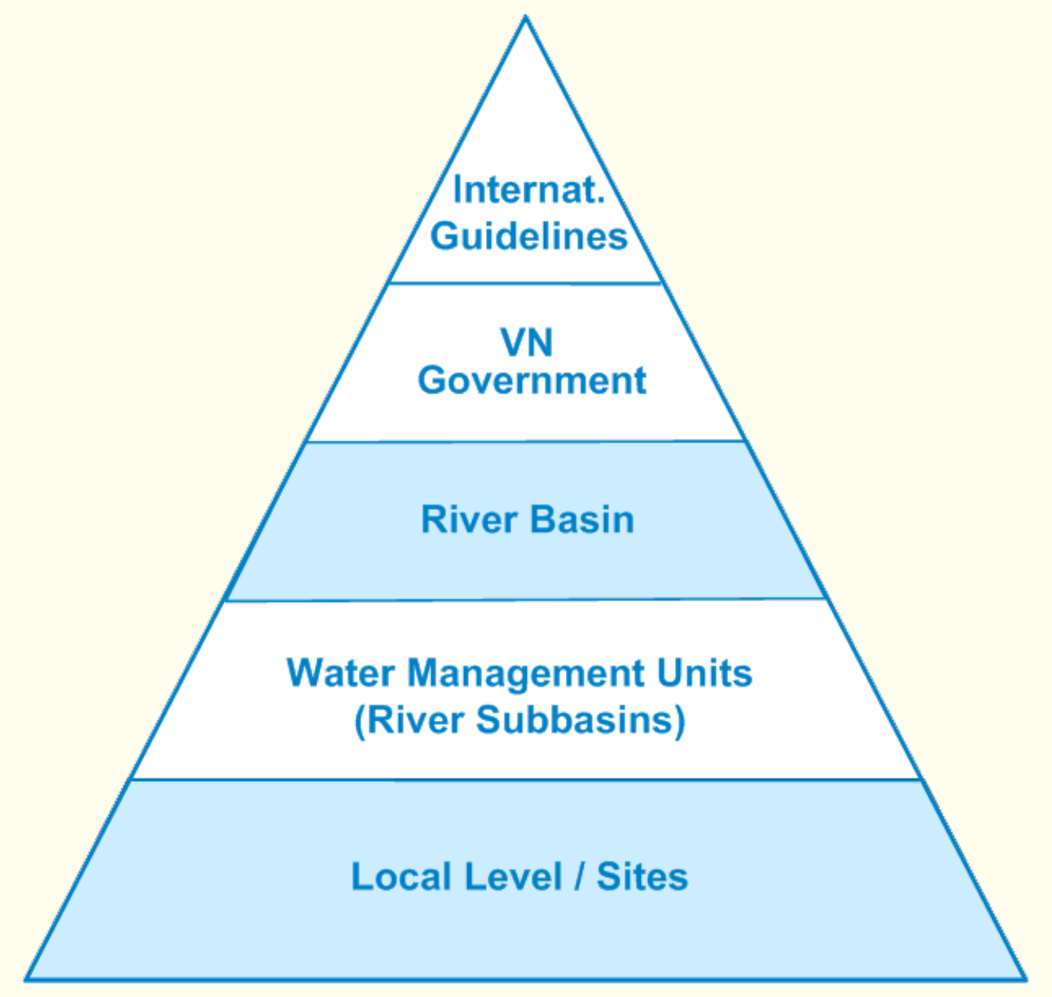
Internat. Guidelines
VN Government
River Basin
Water Management Units (River Subbasins)
Local Level / Sites

Figure 2. Pyramid of IWRM planning levels

The Planning and Decision Support Tools (PDST) consist of three main tools (cf. fig. 3). The tools unify different aspects of water related issues and enable the user to make sustainable planning decisions on a broad basis. The methodical concept of project IWRM-Vietnam follows the ideas of the European Water Frame Directive (EU-WFD). Available concepts are being adjusted to Vietnamese conditions and standards, new concepts are being added to cope with Vietnam's specific challenges. Such, the PDST contribute to the organizational and structural efforts of the Vietnamese government towards an effective water sector.

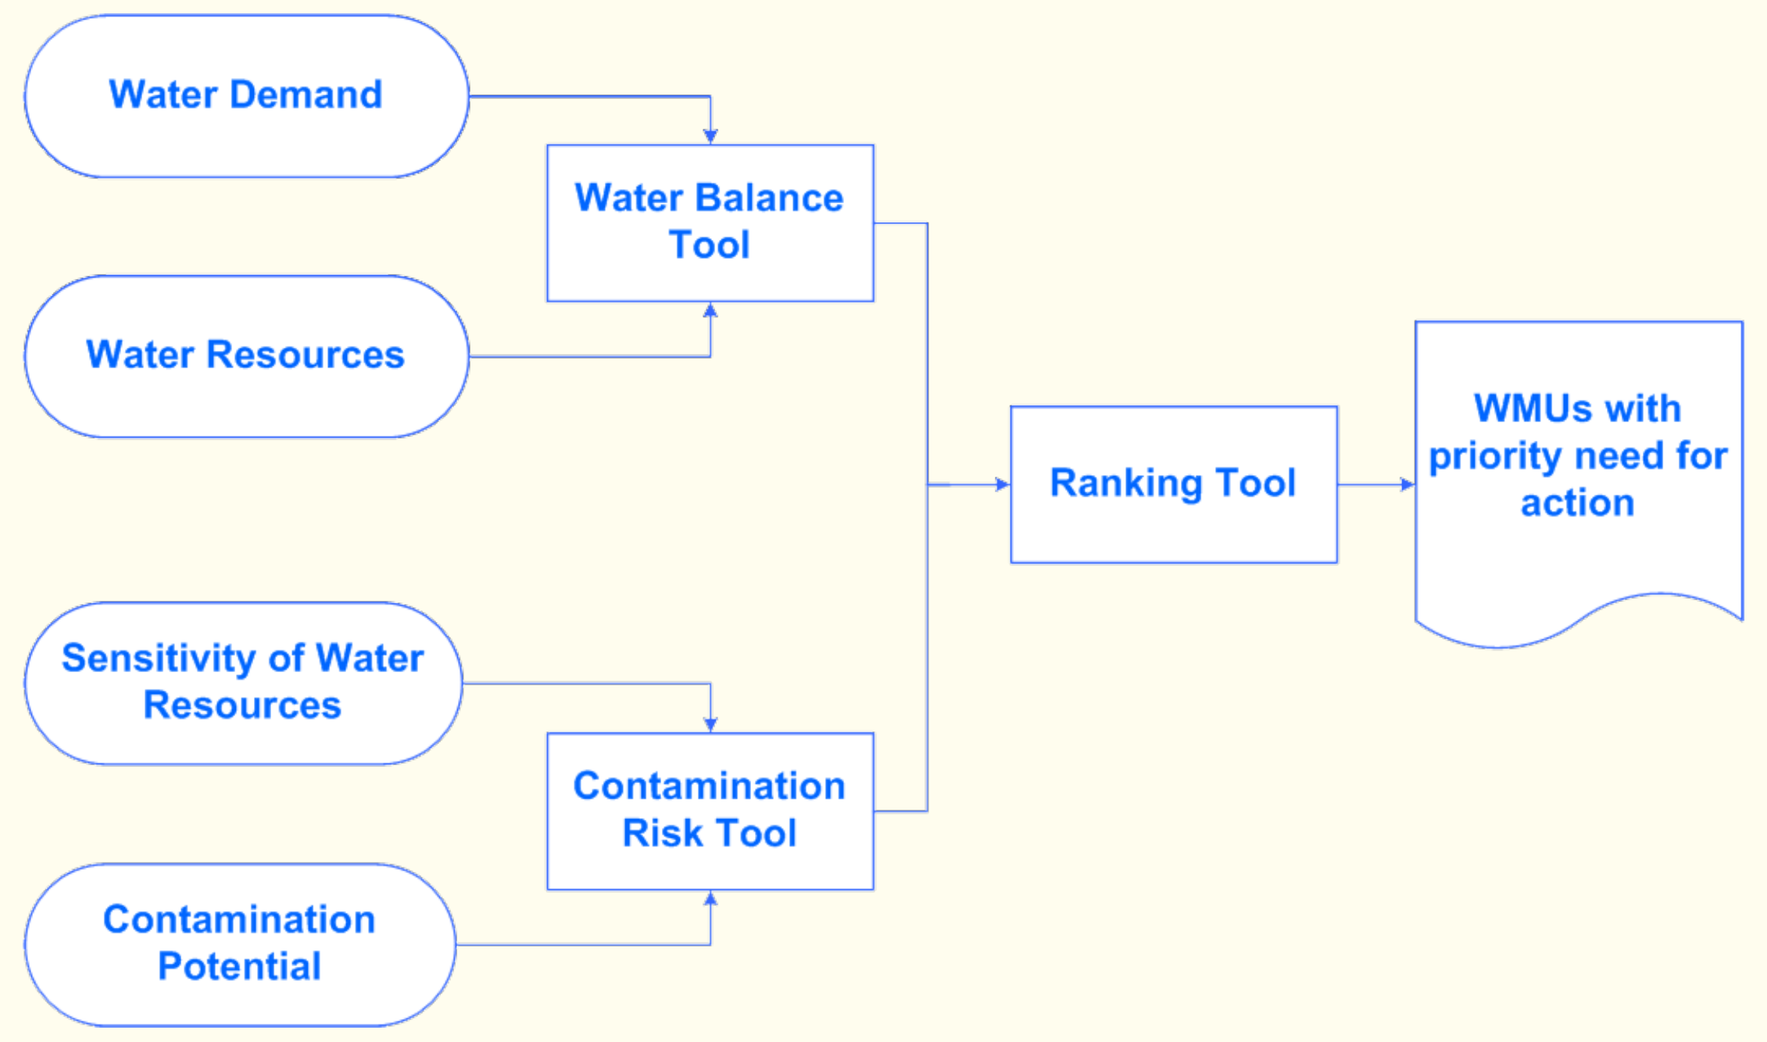
Water Demand
Water Resources
Water Balance Tool
Sensitivity of Water Resources
Contamination Potential
Contamination Risk Tool
Ranking Tool
WMUs with priority need for action

Figure 3. Methodical aggregation: Ellipses symbolize input for PDST (rectangles) to produce evaluation output (last symbol)

Contamination Risk Tool

The Contamination Risk Tool is used to assess contamination risks for water resources (groundwater and surface water) as follows (cf. fig 5-7):

Sensitivity of water resources (SR) + Contamination Potential of polluters (CP)

=

Contamination Risk (CR)

Three possible contamination paths are considered (cf. fig. 4):

1. Infiltration of solute pollutants from diffuse and point sources into groundwater
2. Transport of pollutants from diffuse sources by erosive runoff into surface water
3. Direct discharge of pollutants from point sources into surface water

Figures 5 to 7 show the procedure of the Contamination Risk Tool on the example of path 1 for pollutants from settlements.

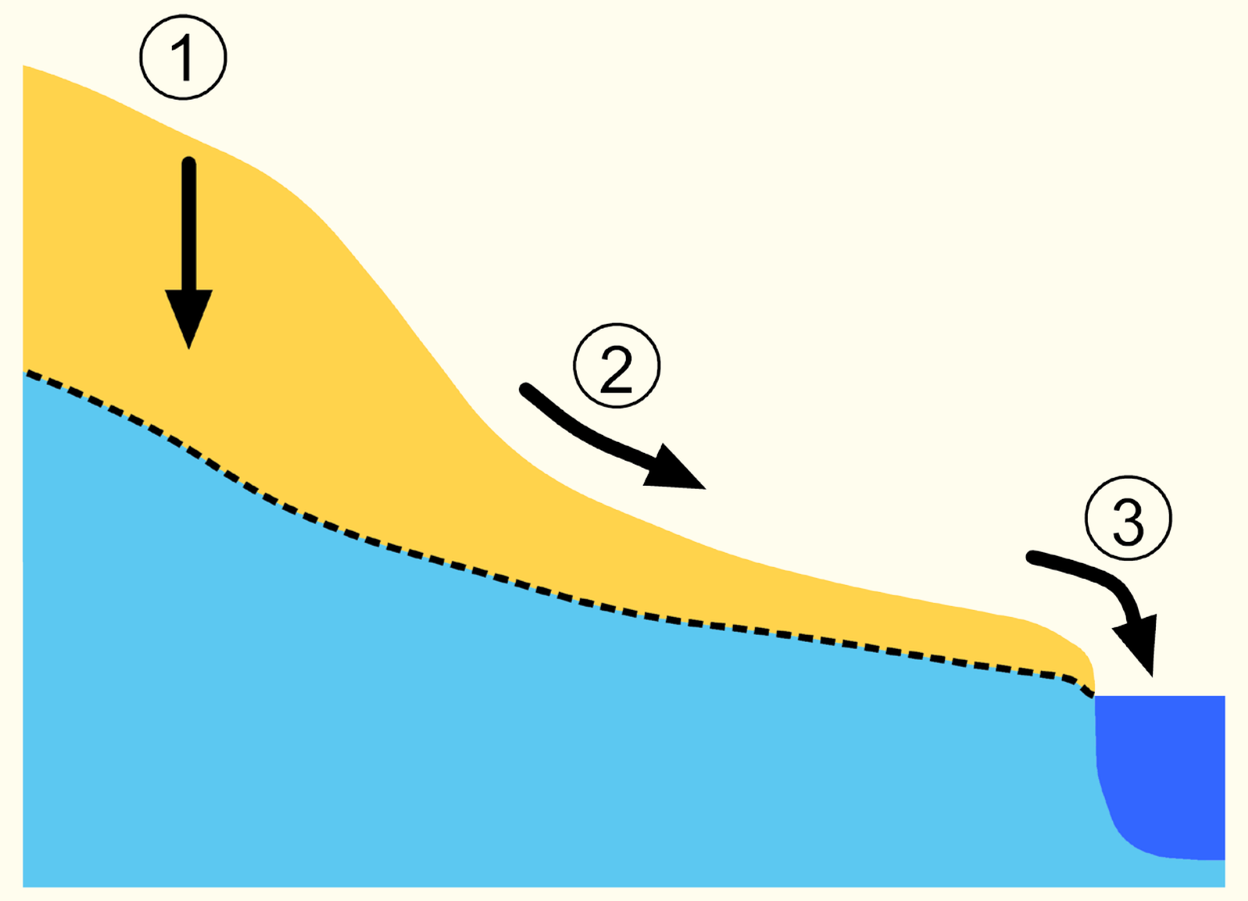
1
2
3

Figure 4. Contamination paths (path 1: infiltration, path 2: erosive runoff, path 3: direct discharge)

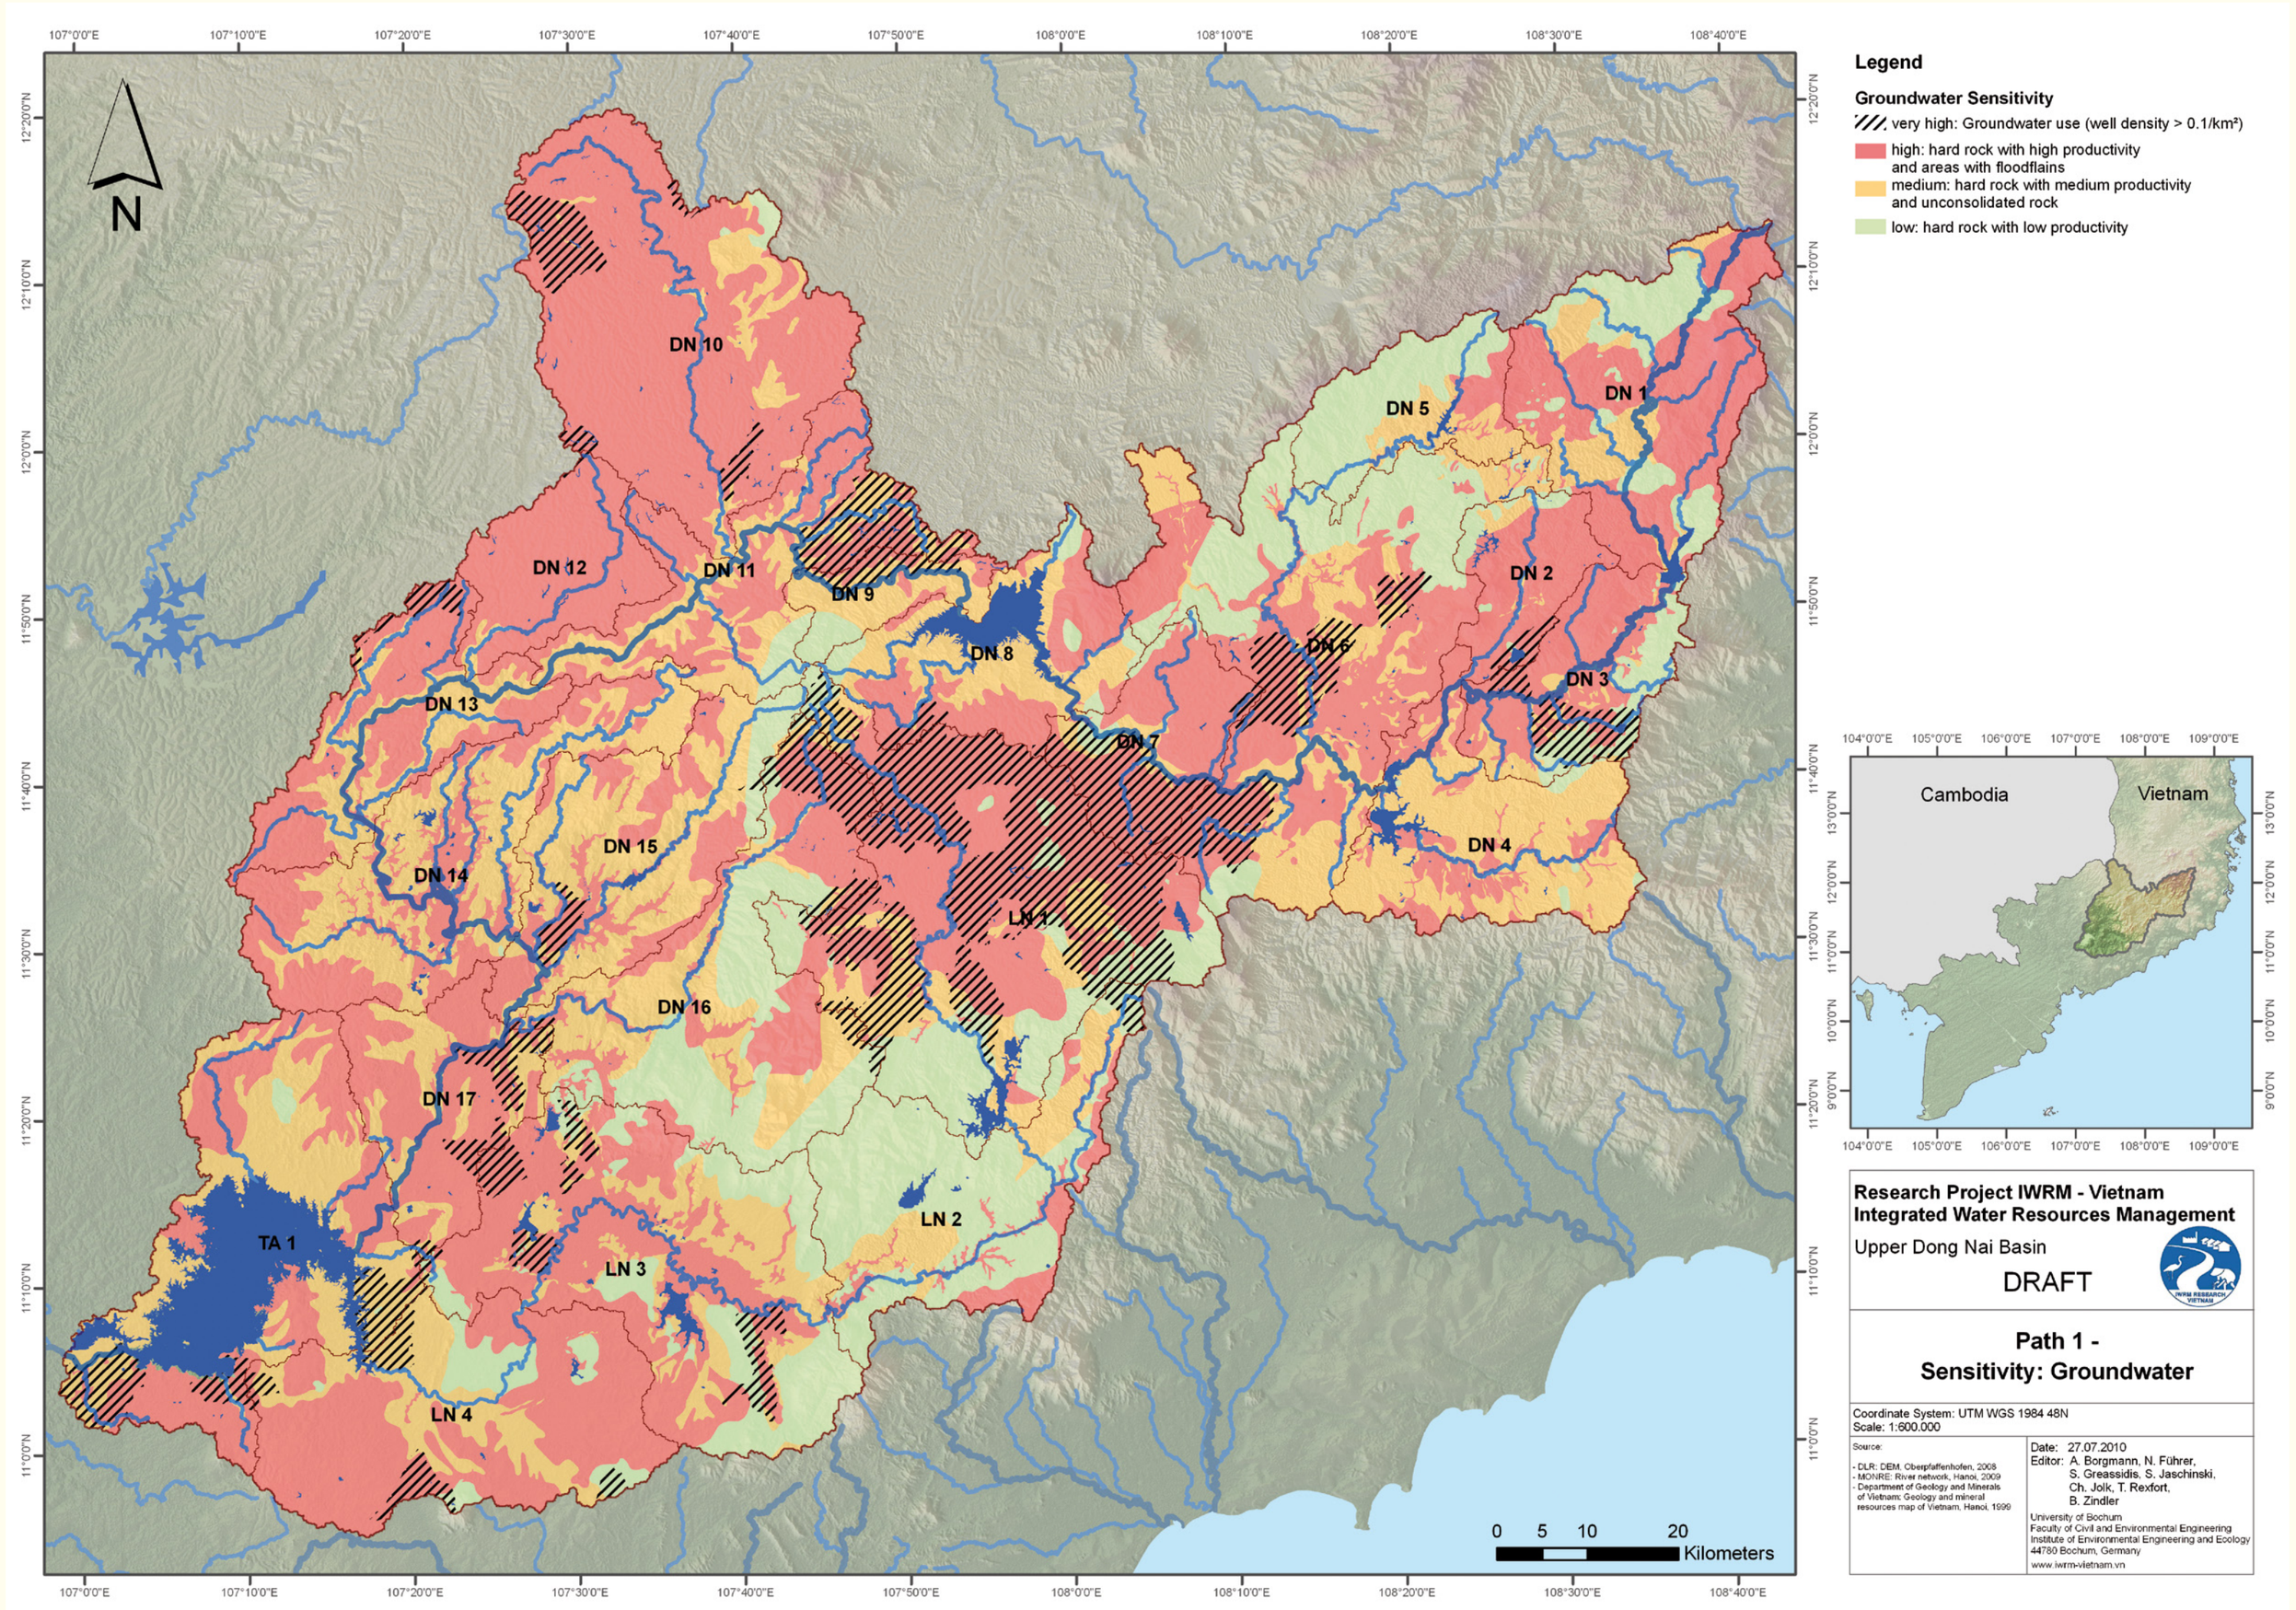
Legend
Groundwater Sensitivity
very high: Groundwater use (well density > 0.1/km²)
high: hard rock with high productivity and areas with floodflains
medium: hard rock with medium productivity and unconsolidated rock
low: hard rock with low productivity
Cambodia
Vietnam
Research Project IWRM - Vietnam
Integrated Water Resources Management
Upper Dong Nai Basin
DRAFT
Path 1 -
Sensitivity: Groundwater
Kilometers

Figure 5. Map of groundwater sensitivity

SR

+

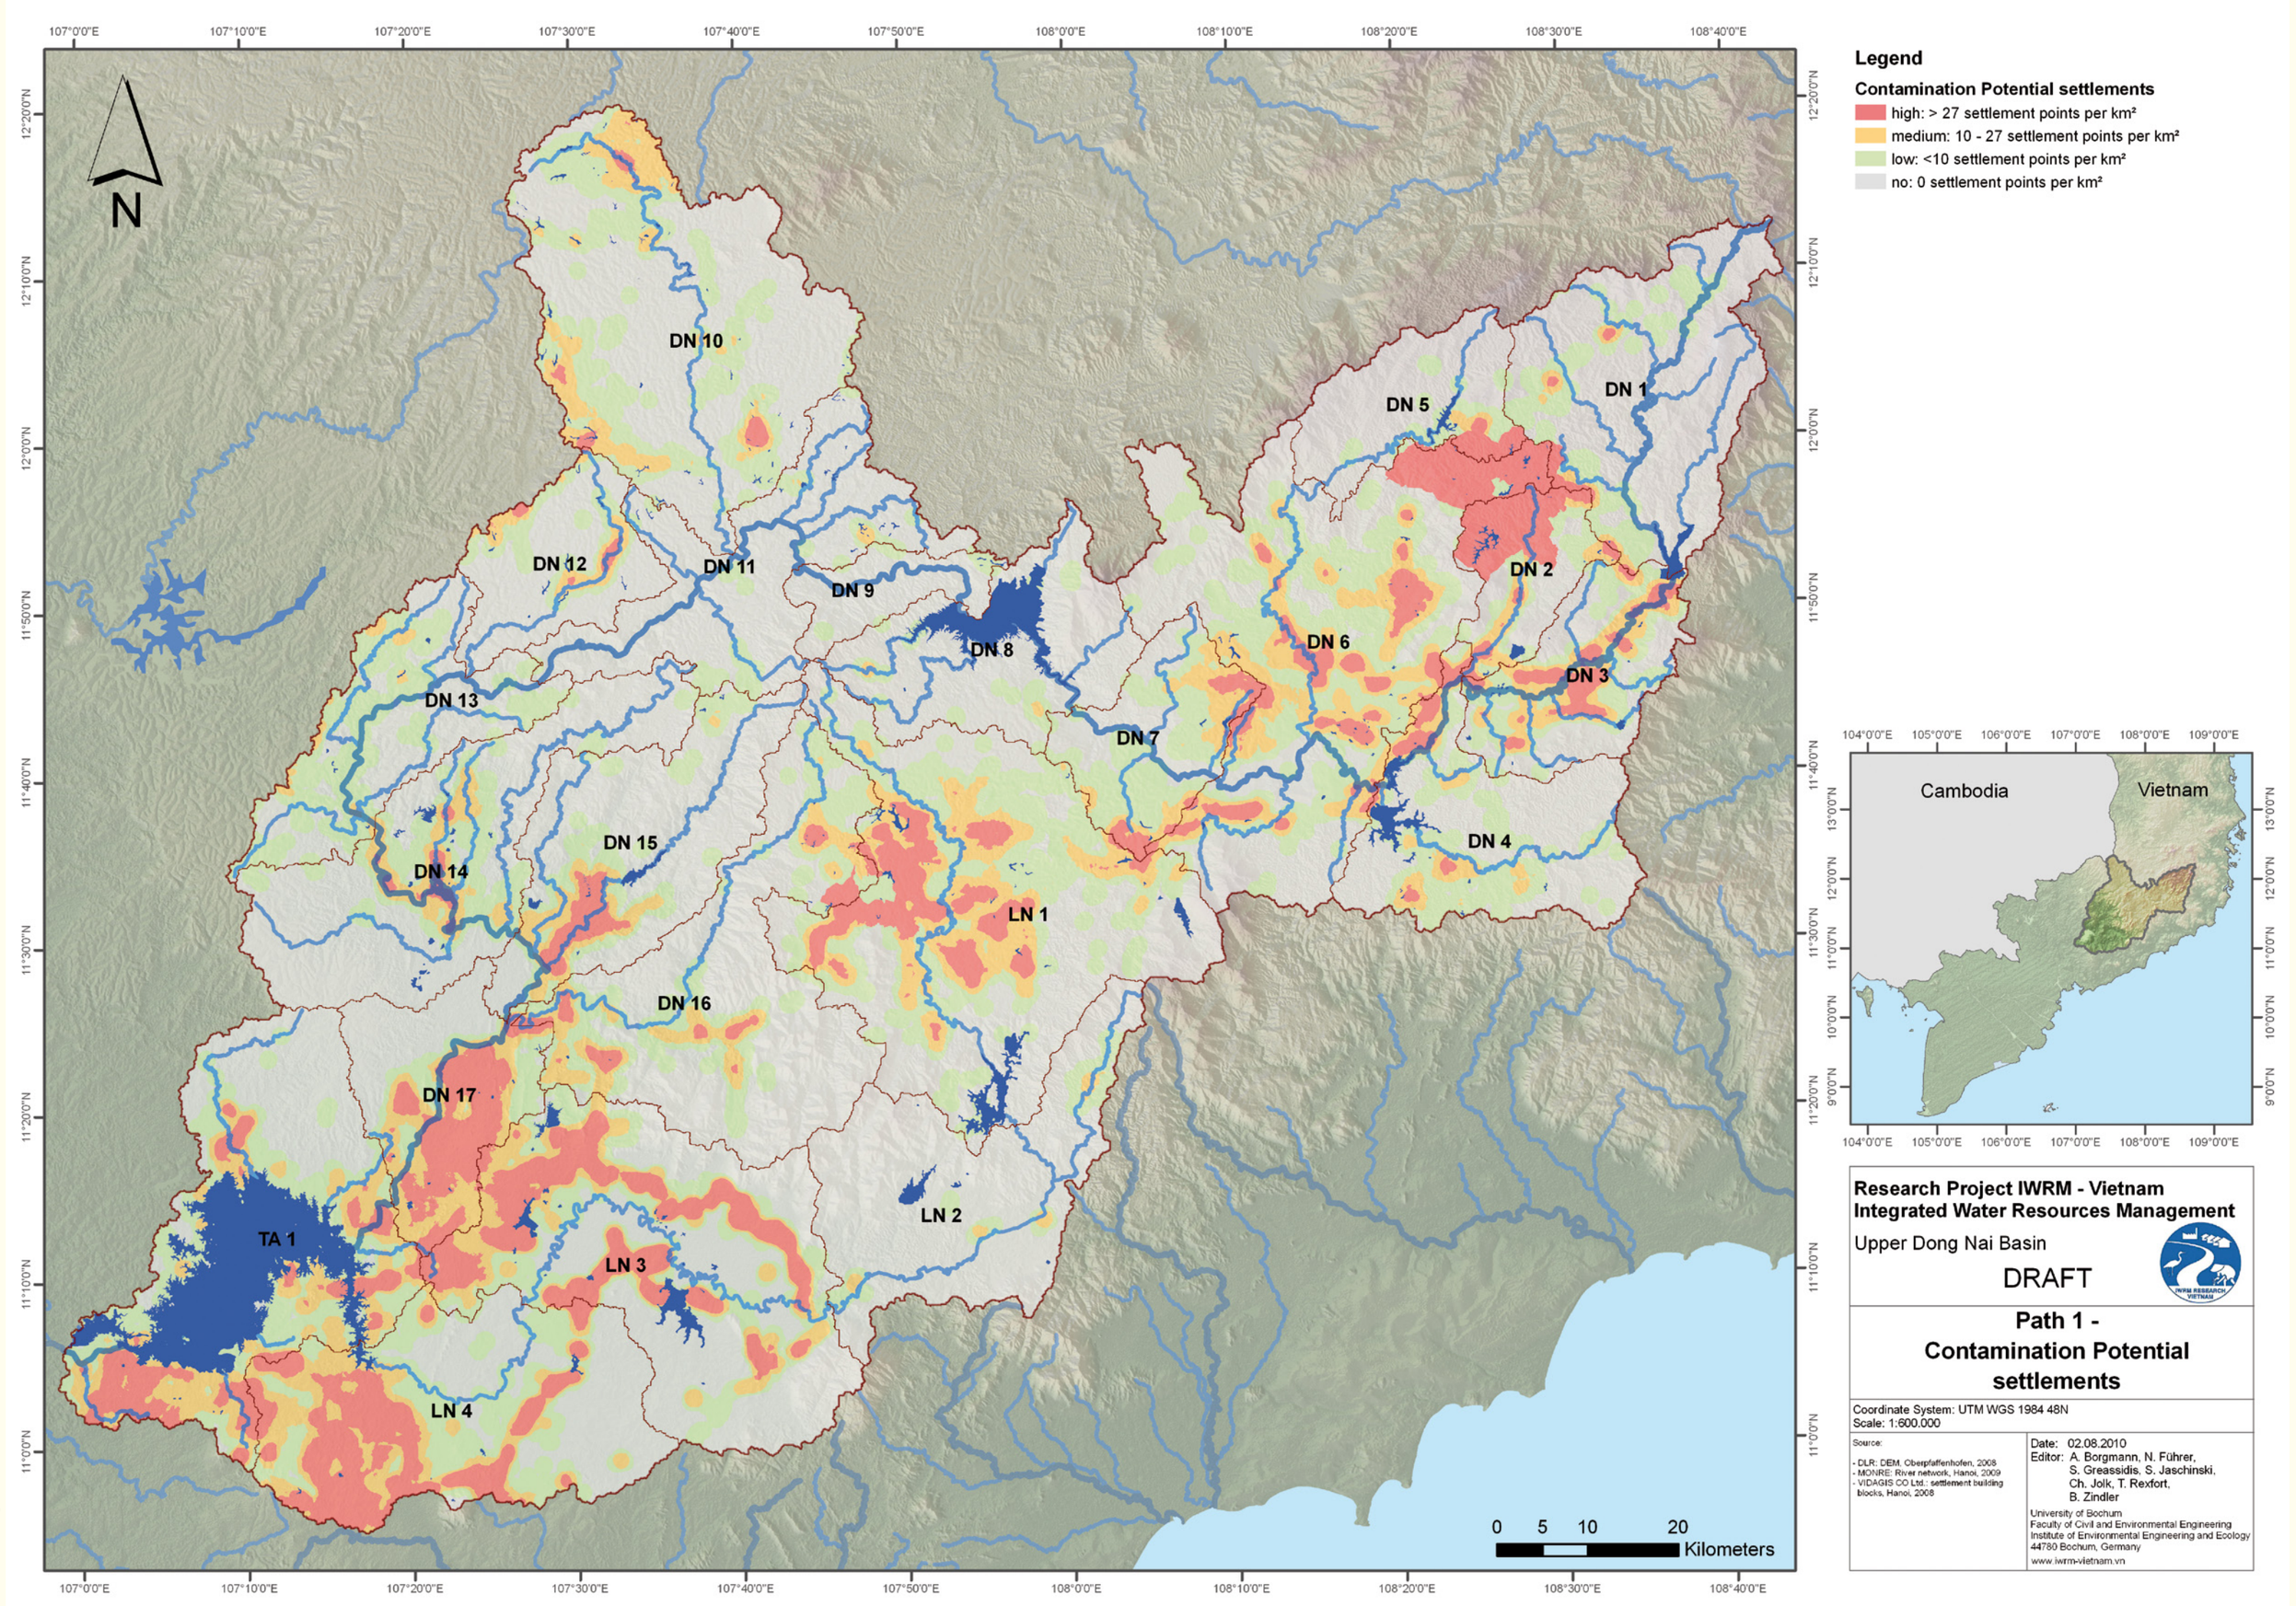
Legend
Contamination Potential settlements
high: > 27 settlement points per km²
medium: 10 - 27 settlement points per km²
low: <10 settlement points per km²
no: 0 settlement points per km²
Cambodia
Vietnam
Research Project IWRM - Vietnam
Integrated Water Resources Management
Upper Dong Nai Basin
DRAFT
Path 1 -
Contamination Potential settlements
Kilometers

Figure 6. Map of contamination potential

CP

=

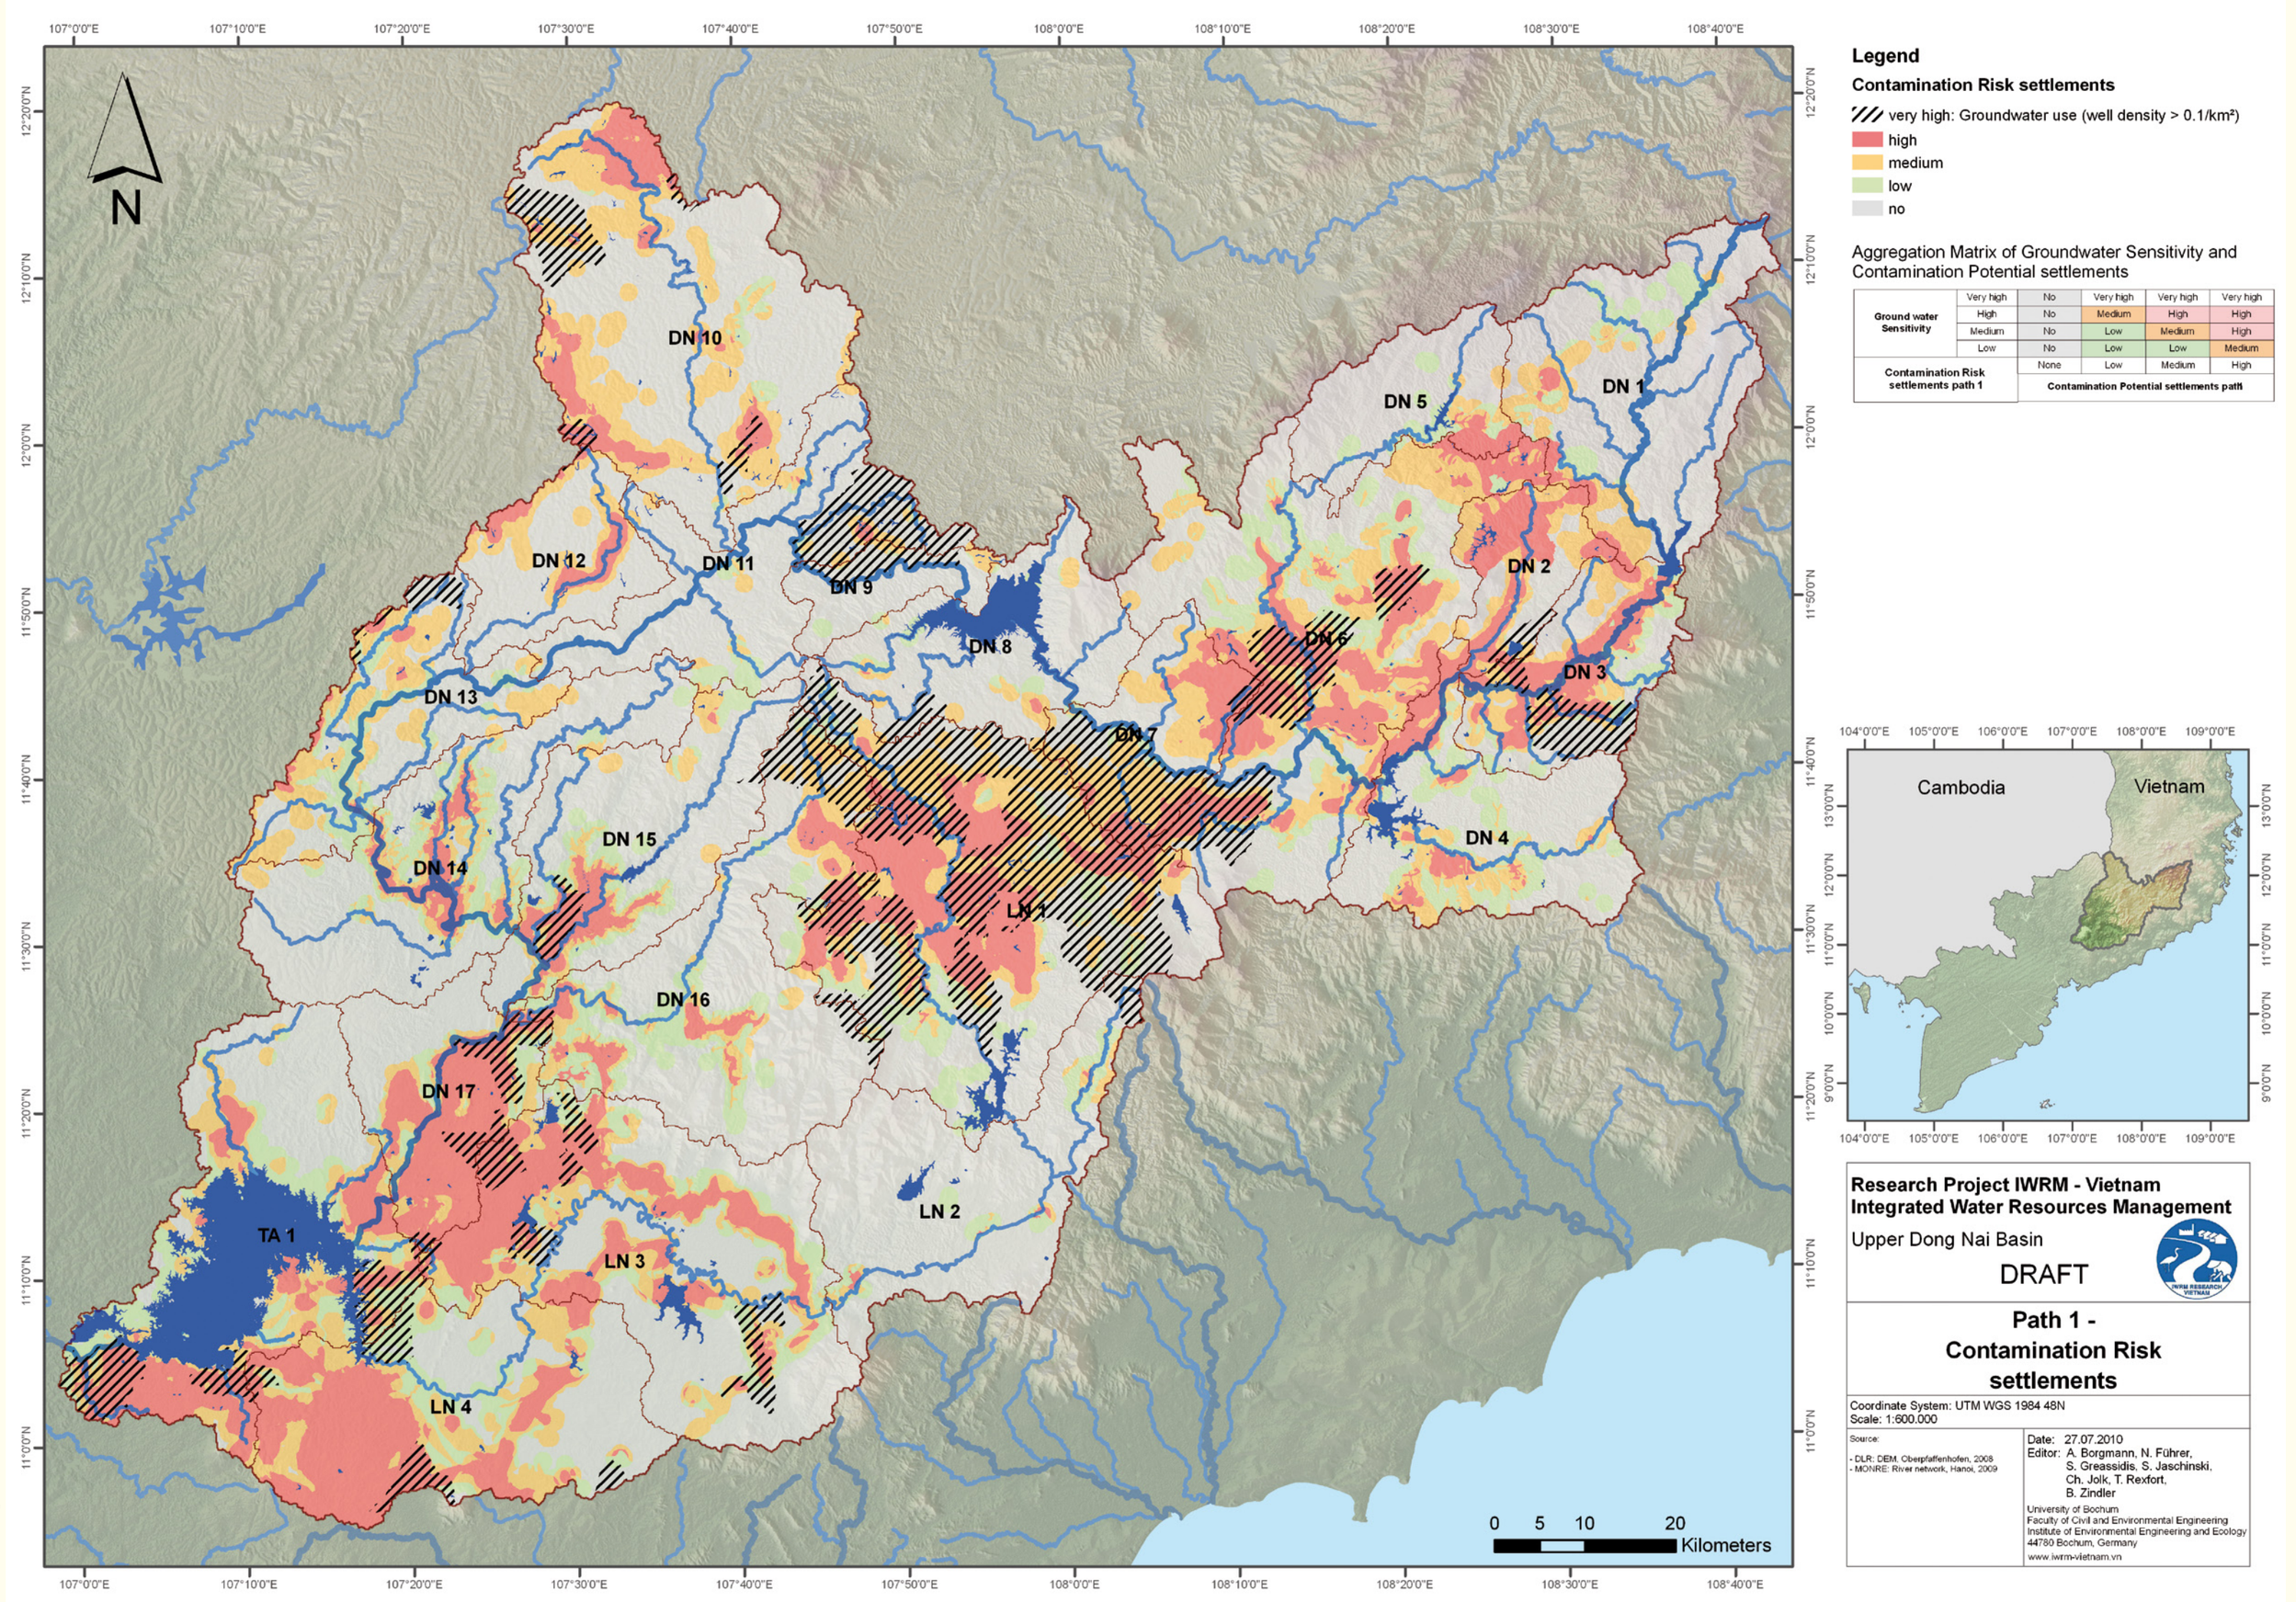
Legend
Contamination Risk settlements
very high: Groundwater use (well density > 0.1/km²)
high
medium
low
no
Aggregation Matrix of Groundwater Sensitivity and Contamination Potential settlements
Cambodia
Vietnam
Research Project IWRM - Vietnam
Integrated Water Resources Management
Upper Dong Nai Basin
DRAFT
Path 1 -
Contamination Risk settlements
Kilometers

Figure 7. Map of contamination risk

CR

Results and Ranking

The Contamination Risk Tool is applicable for all paths and for different causes of contamination potentials under diverse conditions of three dissimilar project regions. Figure 8 shows combinations implemented in the Contamination Risk Tool (cf. figures 10-15 for example photographies).

Contamination paths	Sensitivity	+	Contamination potential	=	Contamination risk
Path 1: Infiltration	Groundwater sensitivity	+	Contamination potential of agriculture	=	Contamination risk of agriculture for groundwater
		+	Contamination potential of settlements	=	Contamination risk of settlements for groundwater
		+	Contamination potential of point sources	=	Contamination risk of point sources for groundwater
Path 2: Erosive runoff	Surface water sensitivity	+	Contamination potential of agriculture	=	Contamination risk of agriculture for surface water
Path 3: Direct discharge	Sensitivity of surface water use	+	Contamination potential of settlements	=	Contamination risk of settlements for surface water
		+	Contamination potential of point sources	=	Contamination risk of point sources for surface water use

Figure 8. Implemented combinations of sensitivity, contamination potential and resulting contamination risk in the Contamination Risk Tool

The example presented in chapter „Contamination Risk Tool" shows the procedure to compile the contamination risk of pollutants from settlements infiltrating into the groundwater in the Upper Dong Nai basin only (cf. fig 5-7).

Figure 5, the groundwater sensitivity, is based on known productivities of aquifers under varying geological situations. Figure 6, the contamination potential, is based on the settlement density of the project region. Figure 7, the resulting map of contamination risk, shows the contamination risk in graded colors from high (red), medium (orange) and low (green) to no risk (grey) areas. According to the blending matrix (cf. legend of fig. 7), areas with high productivity aquifers and settlements with high densities are indicated with high contamination risks. All regions with known groundwater uses via wells are separately considered and classified into very high contamination risks.

The results of all contamination risk assessments for path 1, 2 and 3 are summarized separately within the Ranking Tool for 22 Water Management Units of the project region and are ranked from numbers 1 to 22 where the contamination risk gradually falls from low to high numbers. Figure 9 shows the ranking results for the example given in this presentation.

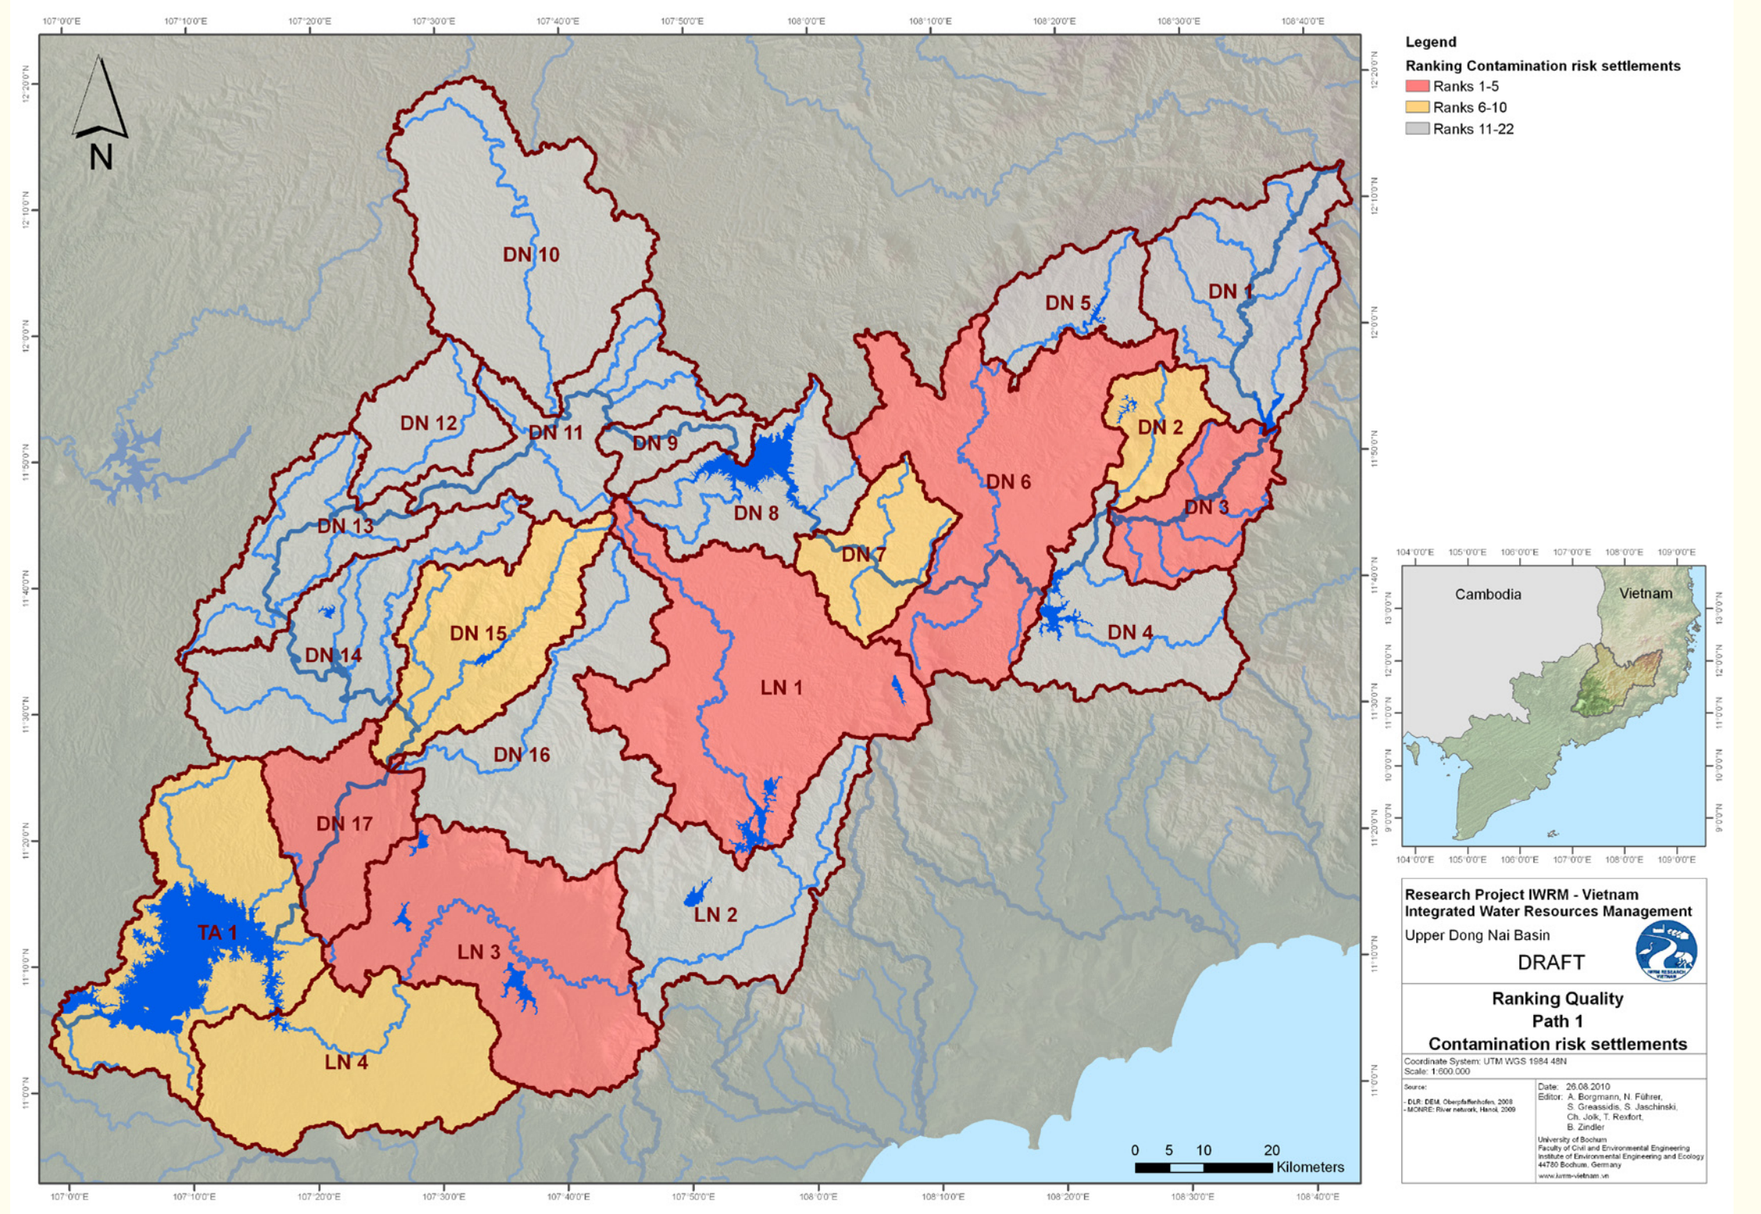
Legend
Ranking Contamination risk settlements
Ranks 1-5
Ranks 6-10
Ranks 11-22
Cambodia
Vietnam
Research Project IWRM - Vietnam
Integrated Water Resources Management
Upper Dong Nai Basin
DRAFT
Ranking Quality
Path 1
Contamination risk settlements
Kilometers

Figure 9. Map of the ranking results for the contamination risk caused by pollutants of settlements infiltrating into groundwater (path 1)

The aim of the Ranking Tool is to identify WMUs with higher problem intensities and priority need for action. The results enable decision makers to derive sound decisions for measures to improve the water quality on river basin scale.

The general need of measures can be identified on the river basin scale. Thus, in order to take IWRM to the next level, a more detailed investigation of the higher ranking WMUs has to be conducted. The PDST need further research and development to be prepared to meet the pressing challenges on a larger scale in order to give assured advice on possible measures on the Water Management Unit and local planning levels (e.g. implementation of a good agricultural practice, establishment of water protection zone concepts, implementation of a monitoring network, construction of water treatment plants).

The Vietnamese stakeholders will be able to base decisions regarding measures to improve the situation of Water Management Units on the Planning and Decision Support Tools. The close cooperation between the project and the Vietnamese authorities ensures a holistic implementation of the tools and a close interaction between the method and legislative framework in Vietnam. The early participation of the responsible water agency on national level guarantees a sustainable adjustment and a nation-wide transferability of the method to Vietnamese conditions. The overall concept developed by project IWRM-Vietnam is an important step towards the implementation of IWRM principles in Vietnam.

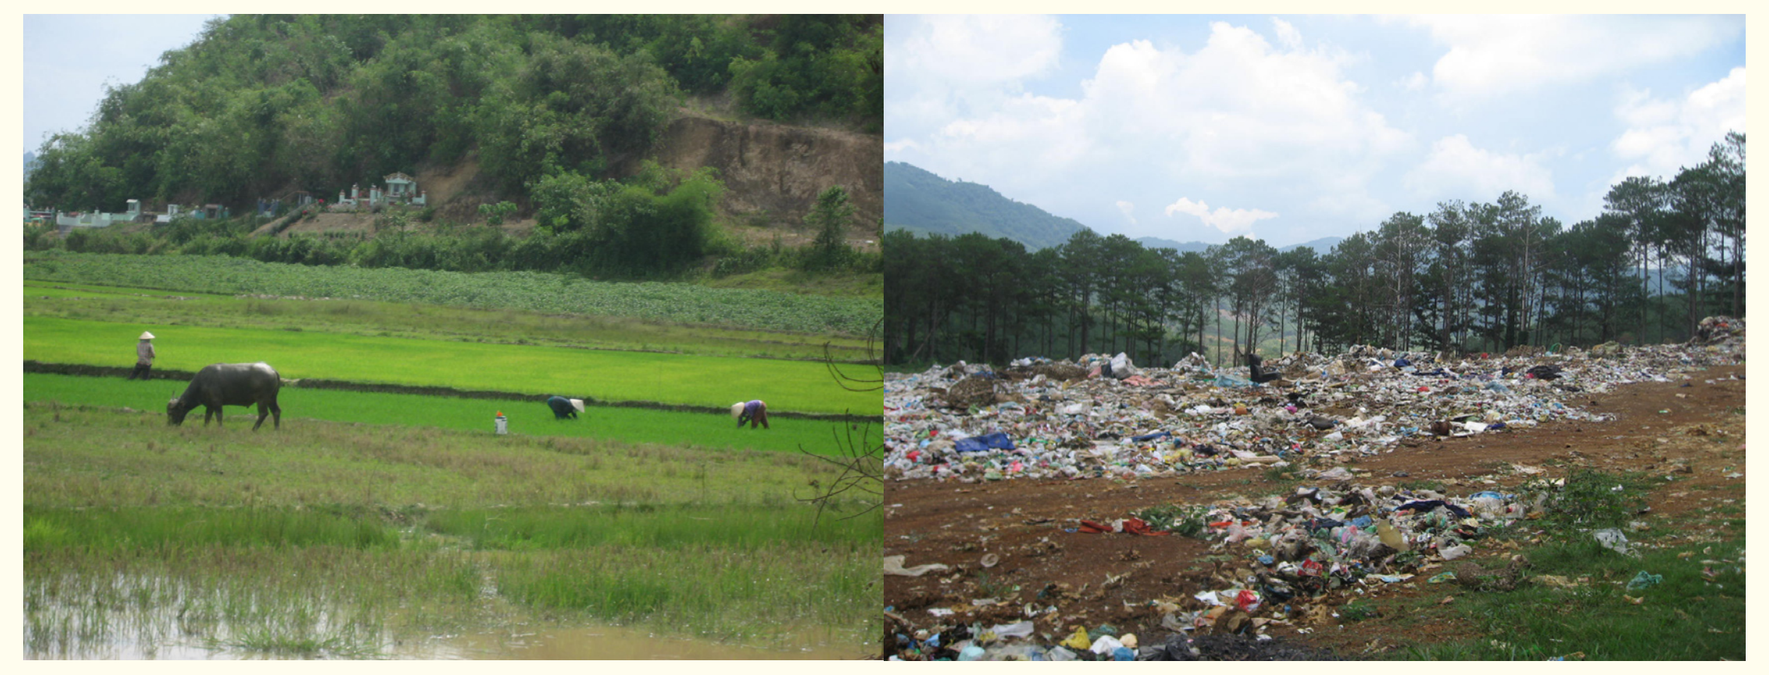

Figure 10, 11. (l. to r.)
Path 1: Infiltration of agricultural pesticides and nutrients into groundwater (rice farming near Da Teh in western Dong Nai river basin);
Path 1: Infiltration of pollutants from a point source into groundwater (unofficial landfill near Di Linh in southeastern Dong Nai river basin)

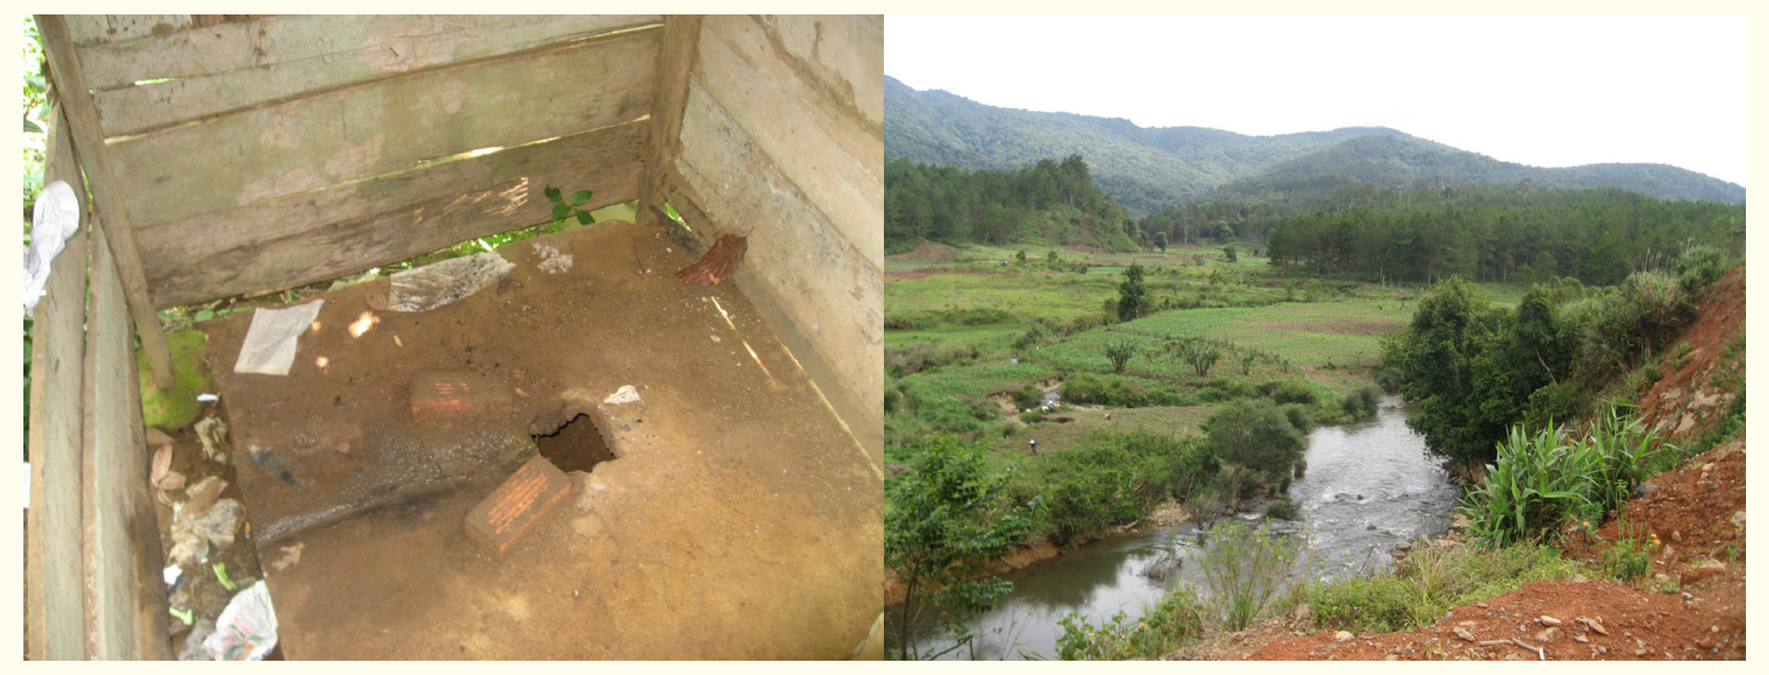

Figure 12, 13. (l. to r.)
Path 1: Infiltration of pollutants from settlements into groundwater (pit latrine in Dak Nong in northwestern Dong Nai river basin);
Path 2: Erosive runoff of agricultural pesticides and nutrients into surface water (annual cultures north of Dalat in Dong Nai river basin)

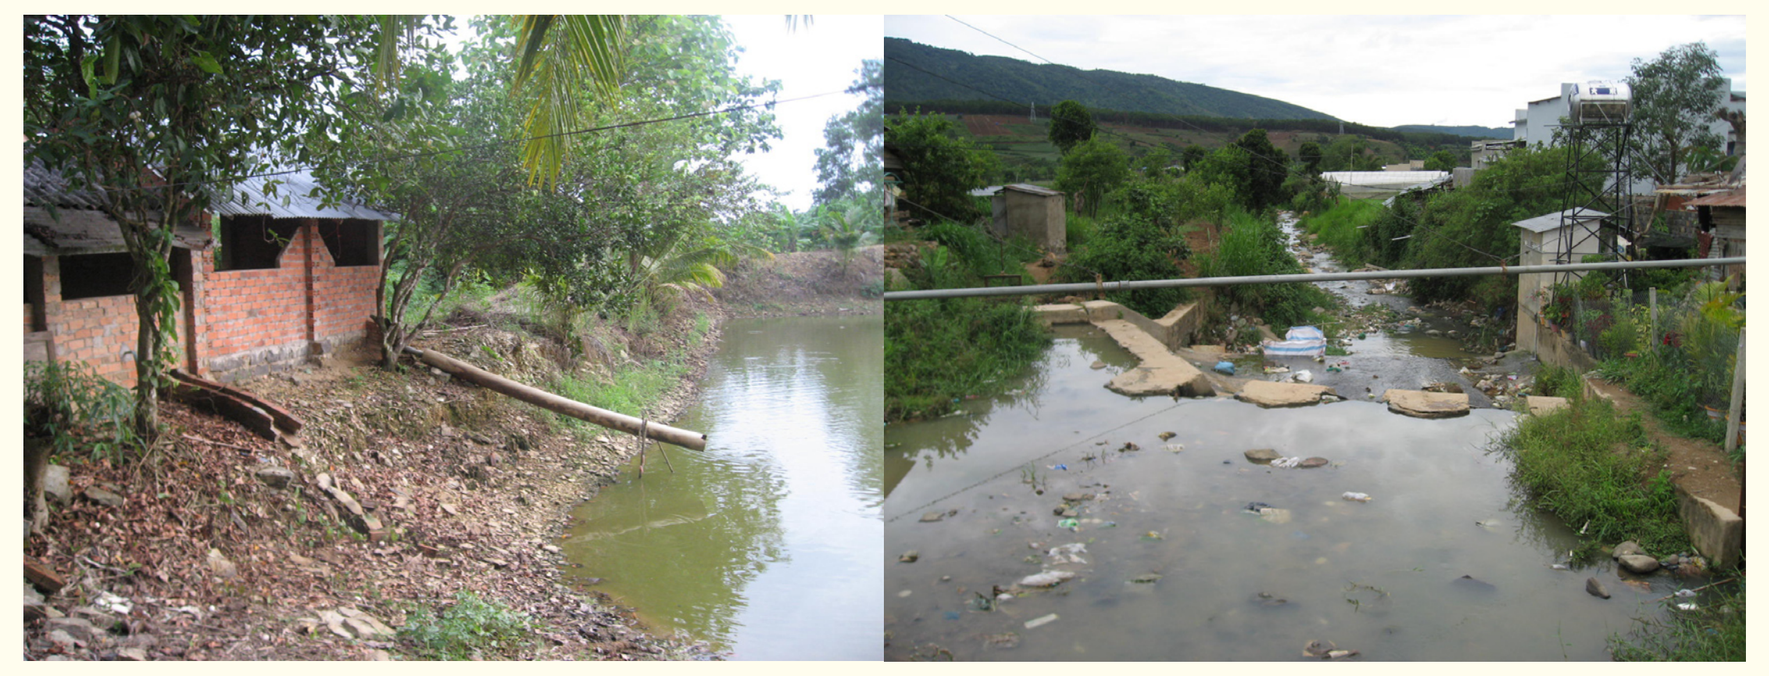

Figure 14, 15. (l. to r.)
Path 3: Direct discharge from a point source into surface water (pigsty near Tri An lake in southern Dong Nai river basin);
Path 3: Direct discharge from settlements into surface water (waste water discharge southeast of Dalat in northern Dong Nai river basin)

References

[1] Cuddihy, W.; Frederiksen, H. (1996): Viet Nam Water Sector Review, World Bank, 1st Ed., Washington, USA.

[2] Government of Vietnam (2008): Decree No. 120/2008/ND-CP on river basin management, Hanoi, Vietnam.

[3] Prime Minister of Vietnam (2007): Decision No. 187/2007/QD-TTg approving the scheme on environmental protection in the basin of the Dong Nai river system up to 2020, Hanoi, Vietnam.

[4] Ministry of Natural Resources and Environment (2006): National Water Resources Strategy towards the Year 2020, 1st Ed., Hanoi, Vietnam.

[5] Agarwal, A.; delos Angeles, M. (2000): Integrated Water Resources Management, Global Water Partnership, 1st Ed., Stockholm, Sweden.

all photographies © by Sylvia Jaschinski, 2009.

eE+E
Environmental Engineering + Ecology
RUB University of Bochum

SPONSORED BY THE
Federal Ministry of Education and Research

IWRM RESEARCH VIETNAM

RUB